Cutaneous Responses in HER2+ Metastatic Breast Cancer in Phase 1b Study of Tucatinib (ONT-380), an Oral HER2 Specific Inhibitor in Combination with Capecitabine and/or Trastuzumab in Third Line or Later Treatment

A.K. Conlin[1], V.F. Borges[2], N.M. Moxon[1], L.N. Walker[3], S.L. Moulder[4]

1. Providence Cancer Center, Portland OR, 2.University of Colorado, Aurora CO, 3.Cascadian Therapeutics, Seattle, WA, 4. MD Anderson Cancer Center, Houston, TX, USA

Background

- Cutaneous metastases in breast cancer are a common and very morbid development in women with metastatic breast cancer (MBC).
- Skin metastases have been estimated to develop in 24% of patients (pts) with MBC, perhaps higher if HER2 overexpressing.[1-2]
- It has been hypothesized that skin may represent a sanctuary site like CNS, for HER2+ patients.[3]
- There have been case reports of patients developing skin only progression on anti-HER2 antibody therapy.

Methods

- This phase 1 study of tucatinib (ONT-380), a potent, oral selective small molecule inhibitor of HER2-neu, dosed in 2 cohorts of 350mg PO BID (8 pts) and 300mg PO BID (52 pts) given with either capecitabine (1000mg/m^2 PO BID 14 days on of 21-day cycle) or trastuzumab (8mg/kg IV load; then 6mg/kg once every 21 days) or the triplet was conducted in women with HER2+ MBC previously treated with a taxane, trastuzumab and T-DM1.
- Prior lapatinib, neratinib, and pertuzumab were allowed. Tumor response was assessed by RECIST 1.1.
- Sixty patients were treated on study and 27 received the triplet of tucatinib with capecitabine and trastuzumab.
- We report on eight patients who had skin noted as site of either measurable or non-measurable disease.

Demographics

Age (range)	36-58	**Site of metastatic disease**			
Race		Skin	8 pts	Liver	1 pt
Caucasian	5 pts	Lymph nodes	5 pts	CNS	2 pts
Hispanic	2 pts	Lung	2 pts	Pleural cavity	1 pt
Black/AA	1 pt	Bone	2 pts		

Prior Therapy

Prior Lines of Therapy	
Median (range)	6 (3-9)
Lapatinib	7 pts
Pertuzumab	6 pts
Radiation to Chest Wall/Skin	7 pts

Results

Treatment of Patients with Skin Lesions

Treatment on Study	**n**
Tucatinib + capecitabine	2
Tucatinib + trastuzumab	1
Tucatinib + capecitabine + trastuzumab	5

Best Overall Response to Therapy

Response Category	**n**
Overall Response	
Complete response	1
Partial response	3
Stable disease	4
Skin Response*	
Complete response	1
Partial response	4
Stable disease	3
Time on Therapy	**Cycles**
Median	7.5
Range	4-12

*Skin PR defined as > 30% reduction of sum of diameters of all target skin lesions from baseline skin; CR defined as disappearance of all skin lesions

Screening — Cycle 10, Day 1

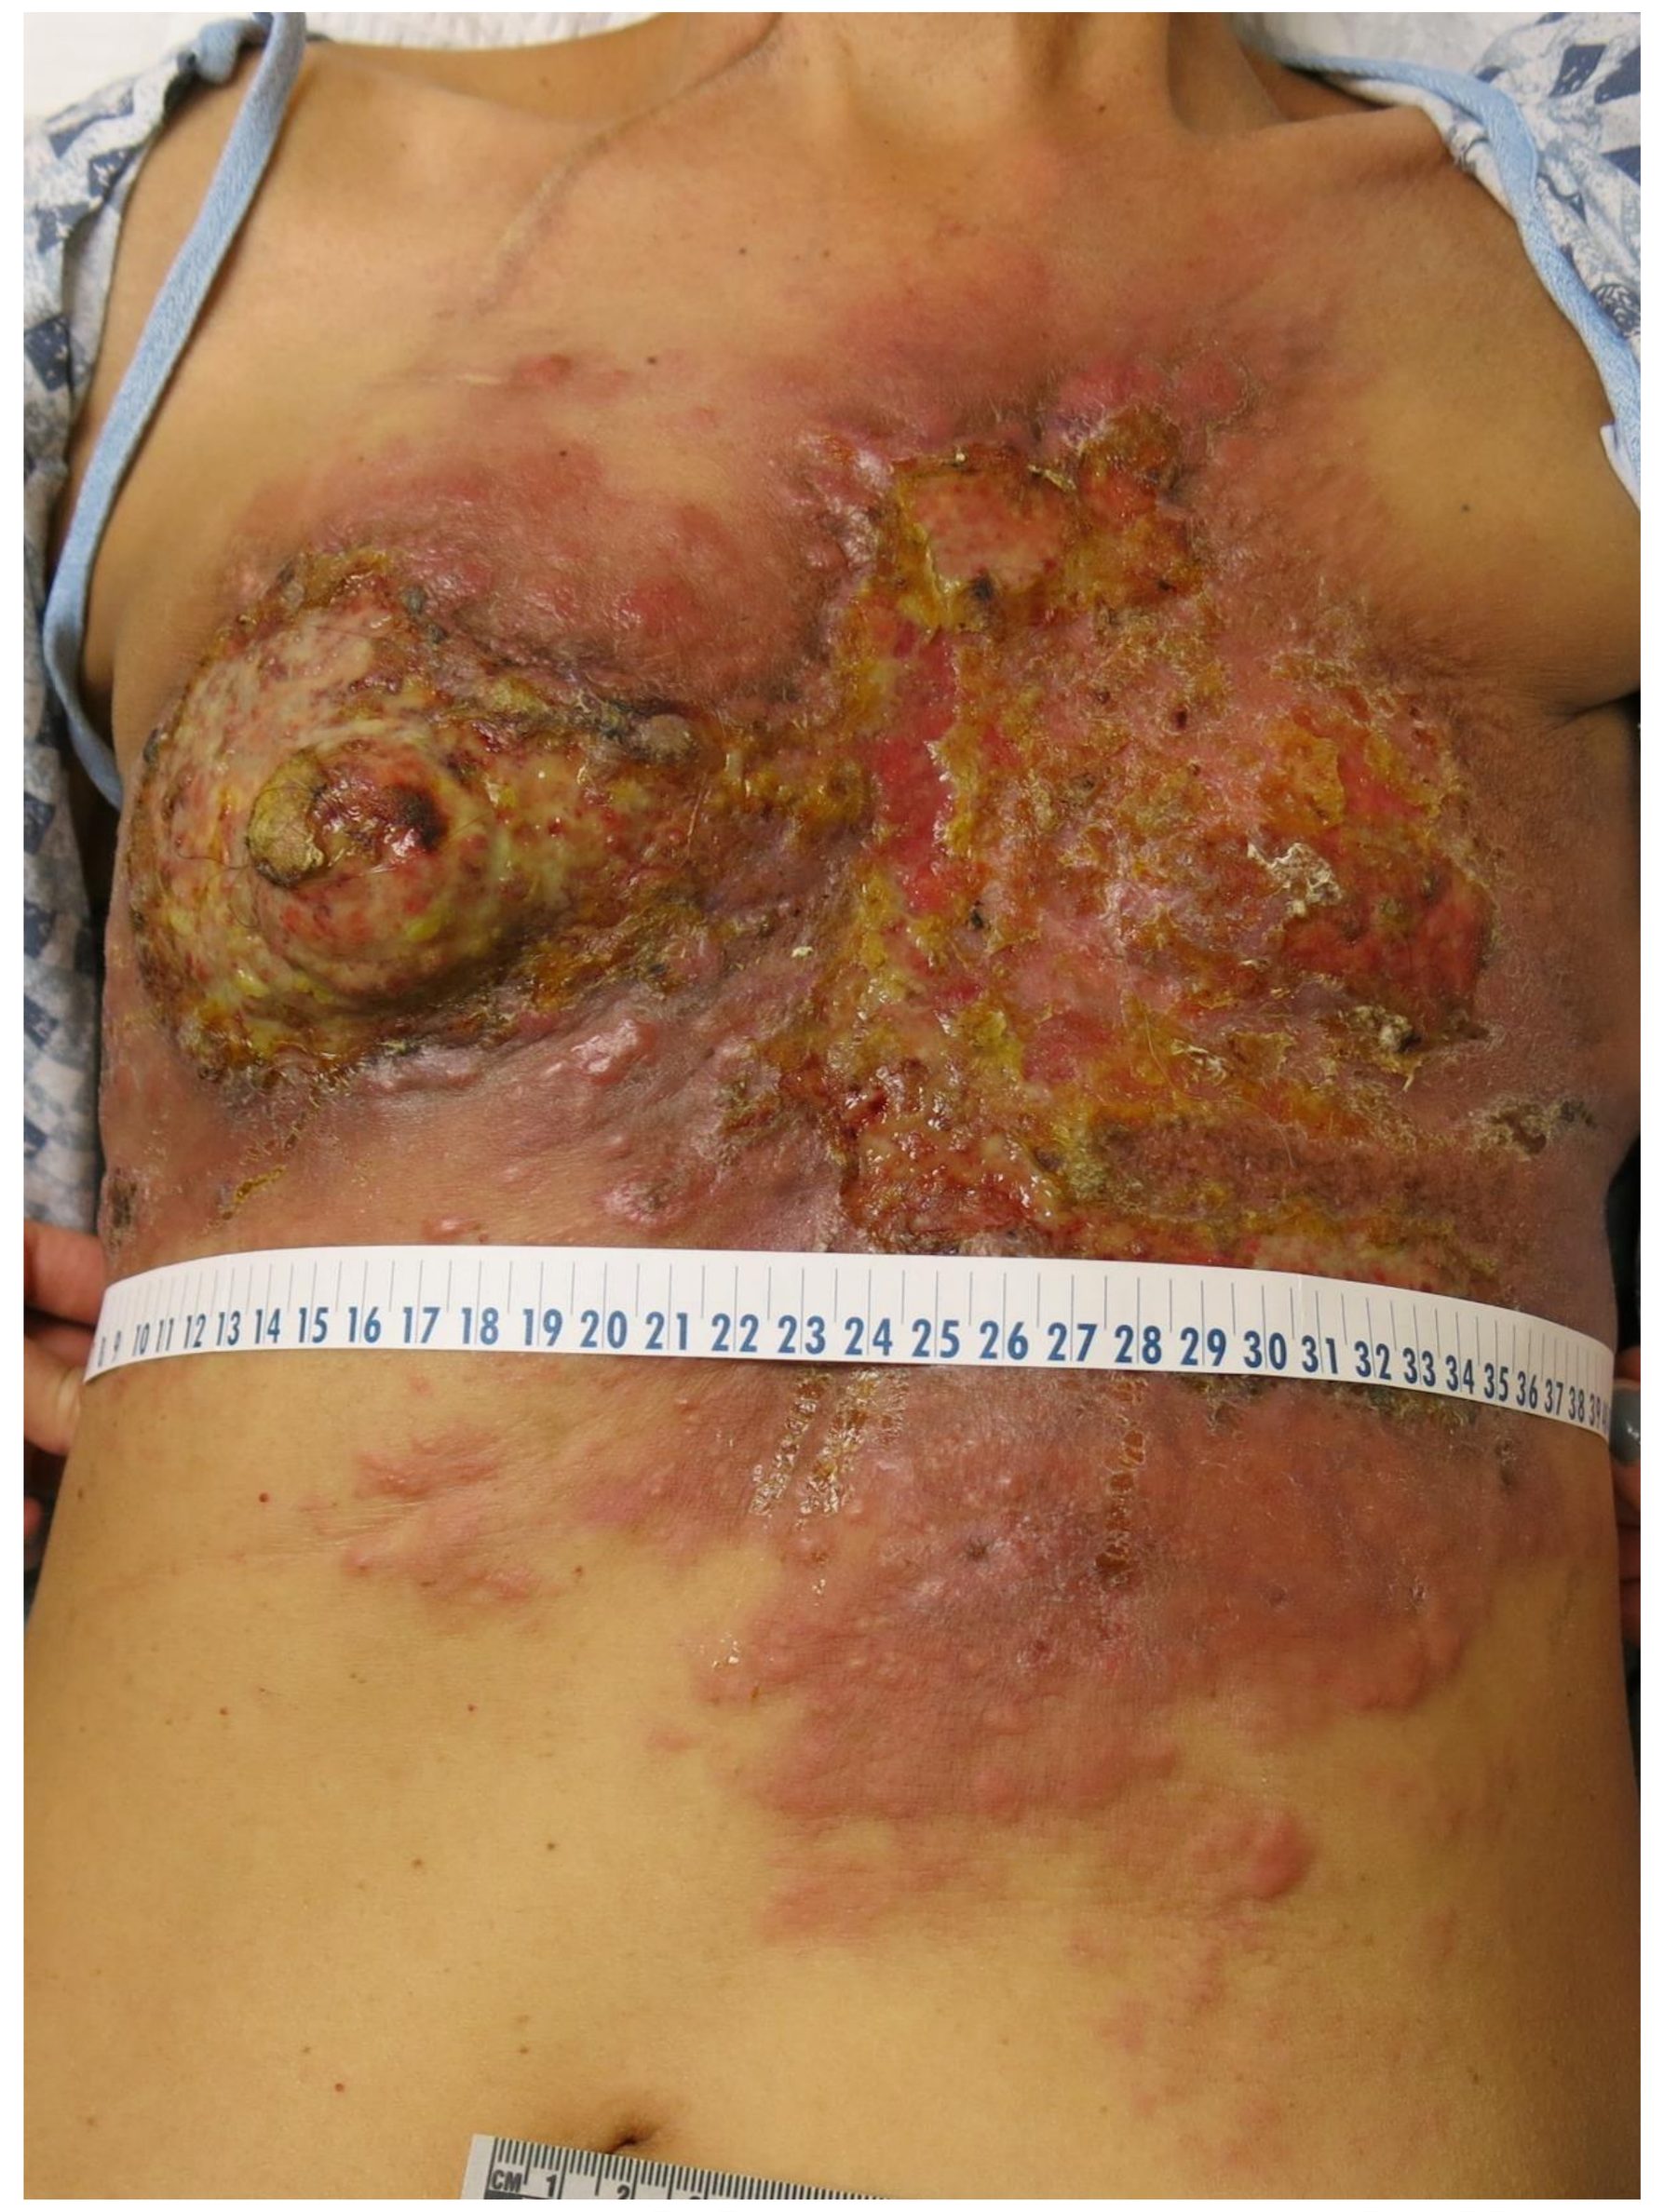

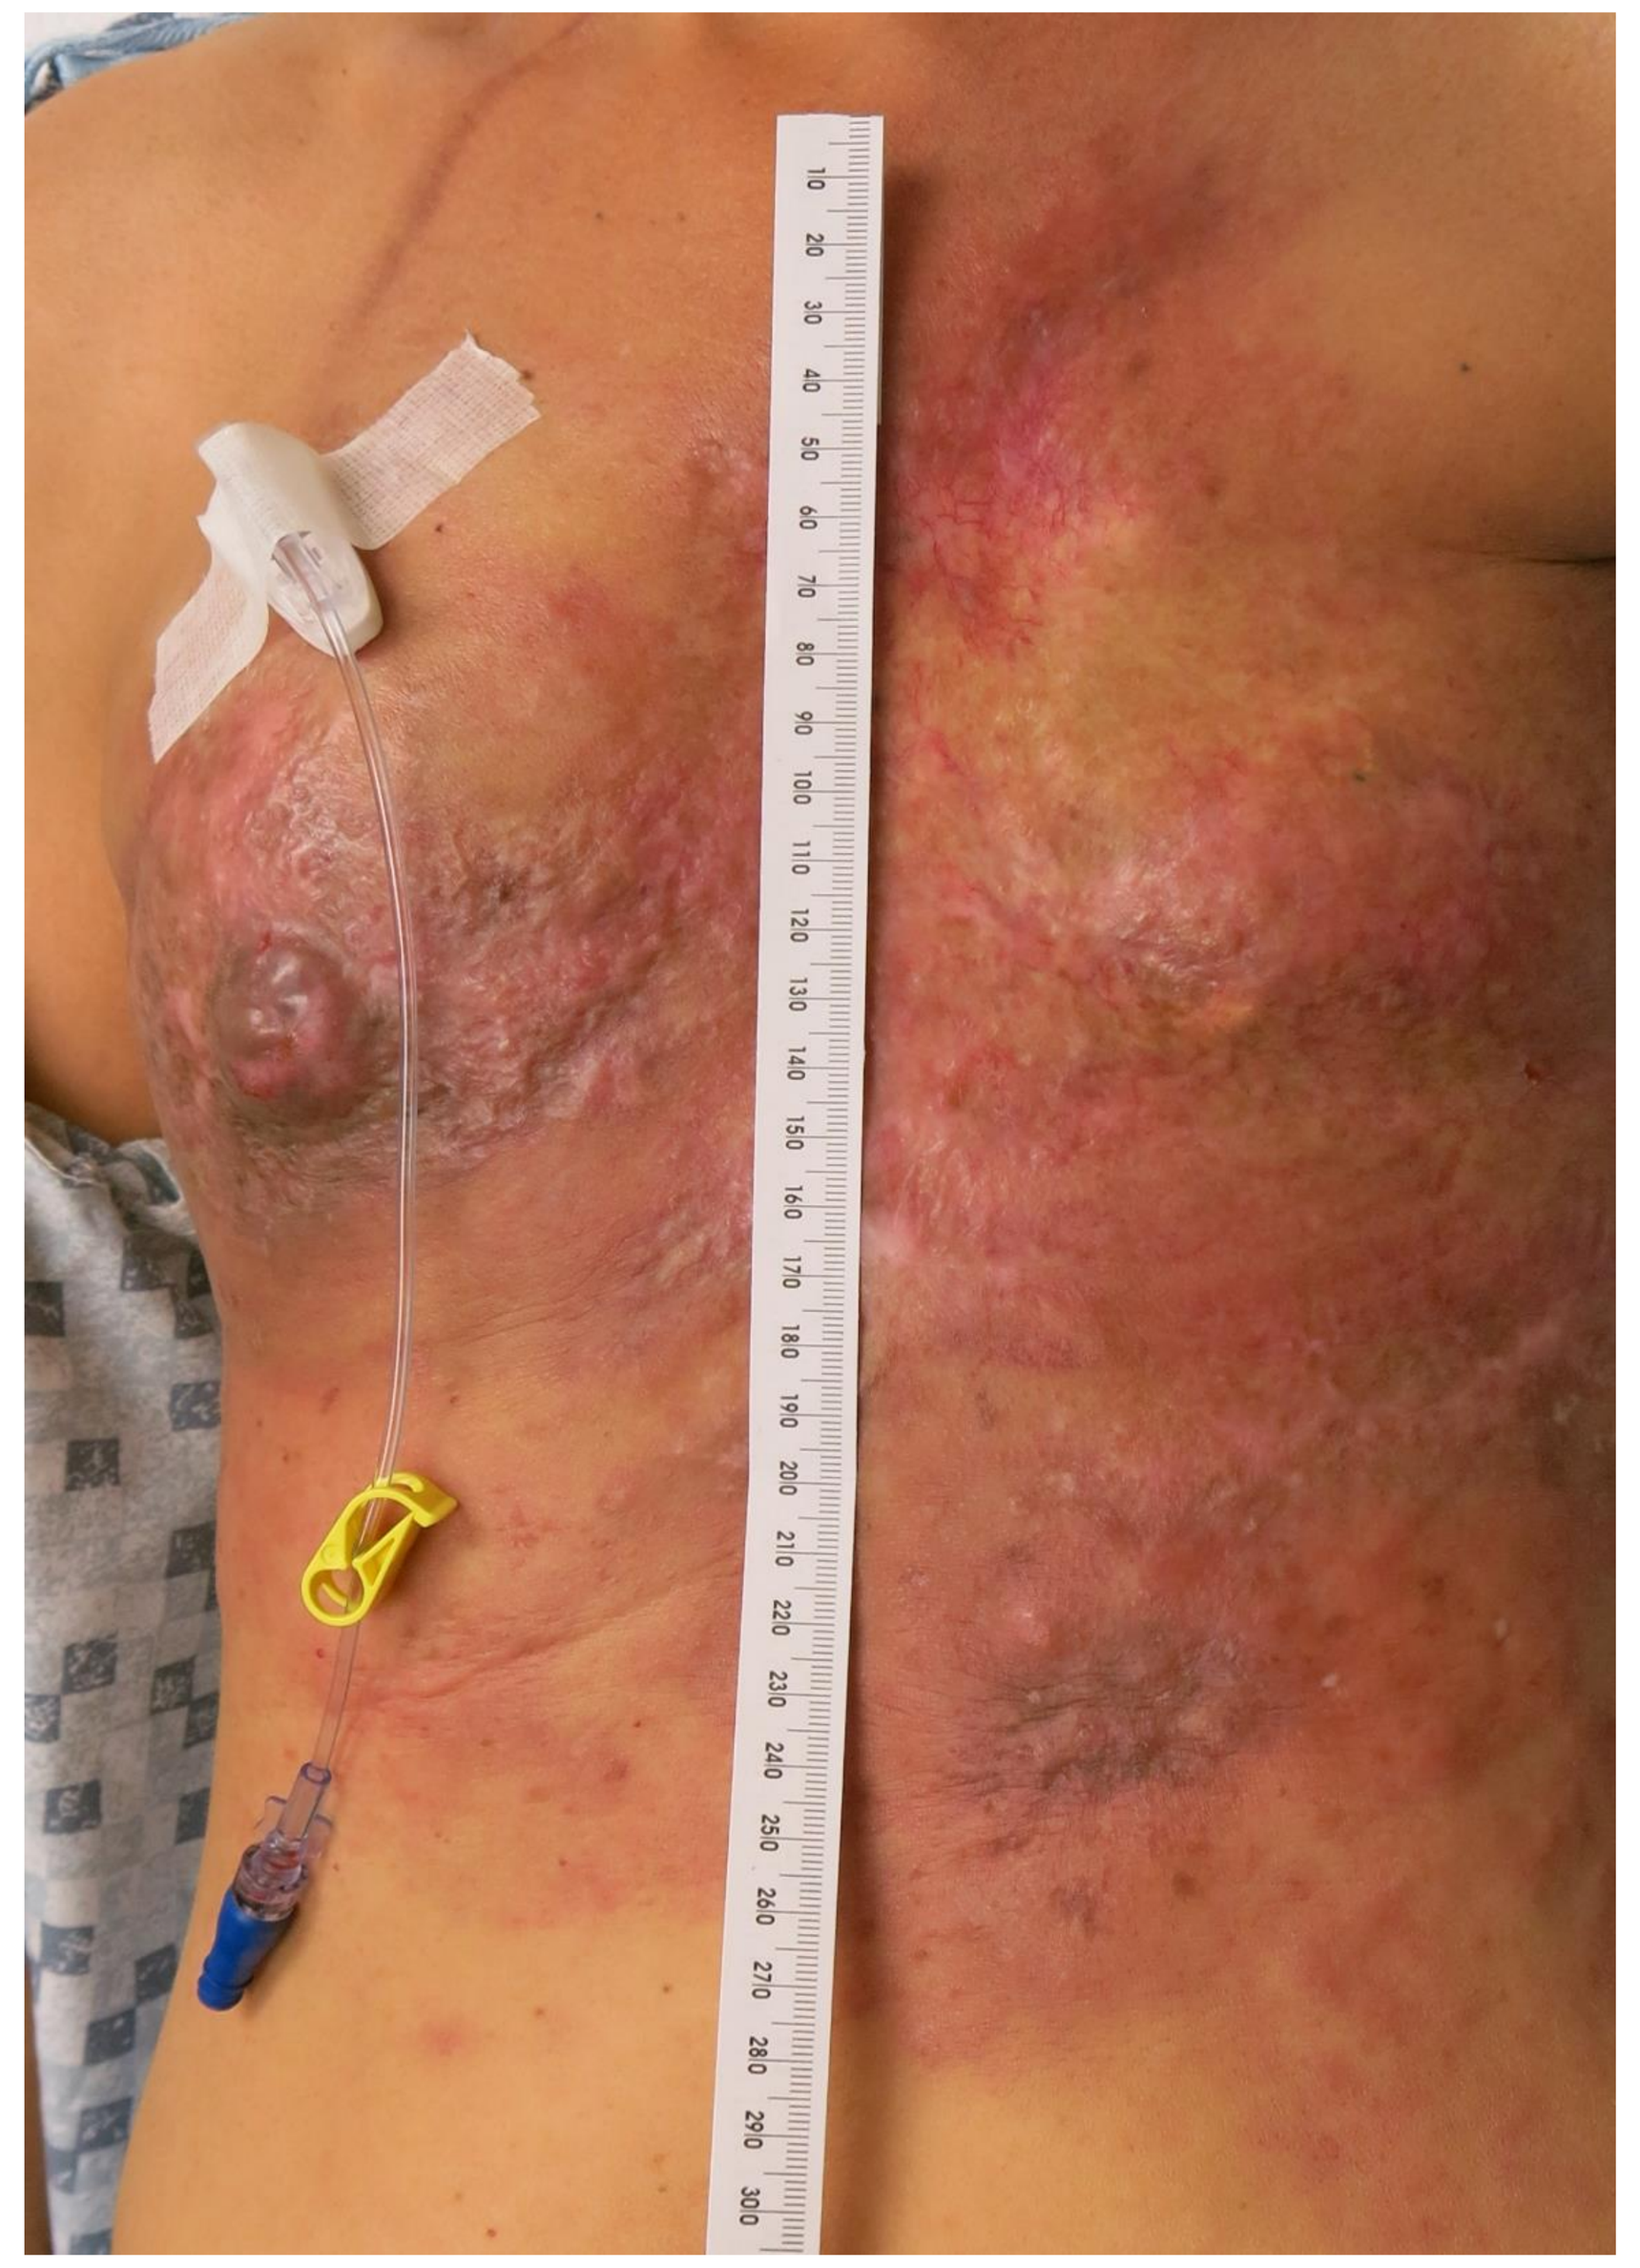

Subject on tucatinib, capecitabine and trastuzumab triplet

Conclusions

- We report on 8 patients with significant response in skin as a disease site while on tucatinib with either capecitabine or trastuzumab or the triplet.
- Tucatinib shows evidence of efficacy in cutaneous metastases, a common and difficult site of disease to control for women with HER2-overexpressing MBC.

References

1. Lookingbill DP et al. J Am Acad Dermatol. 1993; 29: 228-36.
2. Kong JH et al. Oncology. 2011; 81:55-62.
3. Graziano V et al. Cancer Biol Ther 2015; 16(12):1704-9.